668

The Virtual Celiac Symptoms Study: Reported symptoms over 12 weeks in adolescents

Dawn Adams[1], Lisa M. Meckley[2], Jessica R. Marden[3], Sakshi Sethi[3], Elyse Swallow[3], Marilyn Geller[4], Edwin Liu[5], Daniel A. Leffler[2]

[1]Vanderbilt University Medical Center, Nashville, TN, USA; [2]Takeda Development Center Americas, Inc., Cambridge, MA, USA; [3]Analysis Group, Inc., Boston, MA, USA; [4]Celiac Disease Foundation, Woodland Hills, CA, USA; [5]Digestive Health Institute, Children's Hospital Colorado, University of Colorado School of Medicine, Aurora, CO, USA

Presenting author: Edwin Liu (Edwin.Liu@childrenscolorado.org)

Introduction

- Celiac disease (CeD) is an autoimmune condition triggered by gluten exposure.[1]
- CeD symptoms are an important metric in disease activity and significantly affect patients' lives.[2]
- The only management option for CeD is a gluten-free diet (GFD), which has major limitations, and patients following a GFD may continue to experience symptoms due to inadvertent gluten ingestion.[3–7]
- Challenges with adherence to a GFD are common among adolescents.[8]
- An increased understanding of GFD adherence and distinct symptom patterns of different patient populations could help to better inform the management of CeD.
- The Virtual Celiac Symptoms Study (VCSS) is the first observational study to assess daily symptom patterns and impacts of CeD in US adults and adolescents adhering to a GFD.

Objective

- To assess GFD experience and CeD-related symptoms in the adolescent population of the VCSS over 12 weeks.

Methods

Study design

- An observational, prospective study (VCSS; NCT05309330) was conducted in the USA among adults and adolescents diagnosed with CeD.
- Participants self-reported information via a smartphone app regarding:
 - CeD diagnosis, demographics and clinical characteristics
 - occurrence and severity of daily symptoms over 12 weeks via the Celiac Disease Symptom Diary 2.1 (CDSD 2.1©),[9] which included questions regarding the severity of diarrhea, abdominal pain, bloating, nausea and tiredness, and the frequency of vomiting and diarrhea
 - known and suspected inadvertent gluten exposure over 12 weeks.
- Participants were recruited by the Celiac Disease Foundation via digital advertisements (email, social media channels, app push notifications, website advertisements and a study microsite).
- Data collection began on July 25, 2022, and was completed on March 4, 2023.

Inclusion criteria

- English-speaking individuals aged 12 to < 18 years at baseline (for the adolescent cohort) and residing in the USA.
- Diagnosis of CeD for ≥ 1 year (self-reported positive biopsy and/or serology).
- Adherence to a GFD for ≥ 6 months.
- CeD-related symptoms (patient-reported) within the past 3 months.
- Daily access to a smartphone and Internet/Wi-Fi/cellular data.

Exclusion criteria

- Planned or current involvement in any clinical study with an investigational drug, or a surgical procedure or gluten challenge during the 3-month observation period.

Data analysis

- The number of days with core CeD symptoms during the 12-week study period were recorded and a weighted mean number of days with each symptom was determined to account for any days with missing data (weighted mean was calculated as the mean proportion of days with reported symptom(s) across participants weighted by the total study period of 84 days).
- Prevalence of core CeD symptoms during the 12-week study period was stratified by self-reported level of GFD adherence and by irritable bowel syndrome (IBS) status of participants at study initiation.
 - An assessment of the symptom prevalence in participants with and without IBS was of interest because the symptoms of IBS and CeD overlap.
- Participants were included in these analyses if they experienced any symptom at least once during the 12-week study period.
- All measures were analyzed using descriptive statistics with R version 4.0.4. The *p* values were generated using a χ^2 test for categorical variables (or Fisher's exact test when the expected value < 5) and analysis of variance (ANOVA) for continuous variables.

Results

Baseline demographics/clinical characteristics

- Adolescents (12 to < 18 years at enrollment) comprised 142 (29.6%) of the 480 enrolled participants (**Table 1**).
 - The mean (standard deviation; SD) age was 14.5 (1.7) years, mean (SD) time since CeD diagnosis was 5.4 (3.4) years, 66.9% of participants self-identified as female and 97.9% reported to be of White race.
- In total, 16 participants (11.3%) reported having a hospitalization or emergency room visit due to CeD symptoms in the past 12 months, most of whom reported adhering to a GFD and "*rarely eating gluten accidentally*" (**Table 1**).
- The most common self-reported comorbidities at baseline were anxiety (39.4%) and depression (21.1%) (**Table 1**).

Frequency of gluten exposure over the 12-week study period

- The mean (SD) number of days with reported gluten exposure (known or suspected) was 6.7 (8.7) (**Table 2**).

Frequency and prevalence of symptoms over the 12-week study period

- The mean (SD) number of days of reported symptoms was 49.7 (24.4) (**Figure 1**).
 - Of the 65 participants reporting moderate, severe or very severe symptoms, the mean (SD) number of days with these symptoms was 55.4 (22.4); weighted mean 67.7.

Key messages

- **Despite adhering to a gluten-free diet (GFD), adolescent patients with celiac disease (CeD) continued to experience gastrointestinal (GI) symptoms over a 12-week observation period.**
- **This study emphasizes the unmet need for the development of therapies for adolescent patients with CeD on a GFD.**
- **GI symptoms may be appropriate endpoints in future research of CeD therapies.**

Table 1. Baseline demographics and clinical disease characteristics of adolescent participants of the Virtual Celiac Symptoms Study (2022–2023) stratified by level of adherence to a GFD.

	Overall	Self-reported level of adherence to a GFD at baseline: I eat gluten-containing foods regularly	I eat a GFD and rarely eat gluten on purpose	I eat a GFD and rarely eat gluten accidentally	I eat a GFD and never eat gluten accidentally or on purpose	*p* value
Number of participants, n (%)	142 (100)	3 (2.1)	9 (6.3)	111 (78.2)	19 (13.4)	–
Age, years, mean (SD)	14.5 (1.7)	14.3 (2.1)	14.2 (1.9)	14.4 (1.7)	15.2 (1.7)	0.26
White race, n (%)	139 (97.9)	3 (100)	8 (88.9)	109 (98.2)	19 (100)	0.28
BMI, kg/m², mean (SD)	20.9 (4.3)	23.1 (0.4)	21.7 (5.4)	20.8 (4.4)	21.0 (3.4)	0.83
Sex, n (%)						0.75
Female	95 (67.0)	3 (100)	7 (77.8)	72 (64.9)	13 (68.4)	
Male	47 (33.1)	0 (0.0)	2 (22.2)	39 (35.1)	6 (31.6)	
Education, n (%)						0.91
Some HS/HS graduate/some college	79 (55.6)	2 (66.7)	5 (55.6)	60 (54.1)	12 (63.2)	
Elementary/middle school	63 (44.4)	1 (33.3)	4 (44.4)	51 (46.0)	7 (36.8)	
Time since CeD diagnosis, years, mean (SD)	5.4 (3.4)	2.2 (1.6)	3.5 (1.6)	5.8 (3.4)	4.8 (3.6)	0.05
Time spent following a GFD, years, mean (SD)	5.3 (3.4)	2.5 (2.2)	2.8 (1.4)	5.6 (3.4)	4.7 (3.5)	0.03
Comorbidities, n (%)						
At least one comorbidity	102 (71.8)	3 (100)	8 (88.9)	76 (68.5)	15 (79.0)	0.43
Anxiety	56 (39.4)	2 (66.7)	4 (44.4)	40 (36.0)	10 (52.6)	0.37
Depression	30 (21.1)	1 (33.3)	4 (44.4)	20 (18.0)	5 (26.3)	0.15
ADHD	24 (17.0)	1 (33.3)	2 (22.2)	17 (15.3)	4 (21.1)	0.52
Anemia	13 (9.2)	0 (0.0)	2 (22.2)	8 (7.2)	3 (15.8)	0.22
GERD	13 (9.2)	1 (33.3)	2 (22.2)	8 (7.2)	2 (10.5)	0.11
IBS	11 (7.8)	1 (33.3)	0 (0.0)	6 (5.4)	4 (21.1)	0.04
Thyroid disease	8 (5.6)	0 (0.0)	0 (0.0)	8 (7.2)	0 (0.0)	0.81
Type I diabetes	3 (2.1)	0 (0.0)	1 (11.1)	2 (1.8)	0 (0.0)	0.28
Failure to thrive as a child or difficulty gaining weight, n (%)	51 (35.9)	1 (33.3)	3 (33.3)	40 (36.0)	7 (36.8)	1.00
Hospitalization or ER visit due to CeD symptoms in the past 12 months, n (%)	16 (11.3)	0 (0.0)	5 (55.6)	10 (9.0)	1 (5.3)	0.004

ADHD, attention-deficit/hyperactivity disorder; BMI, body mass index; CeD, celiac disease; ER, emergency room; GERD, gastroesophageal reflux disease; GFD, gluten-free diet; HS, high school; IBS, irritable bowel syndrome; SD, standard deviation.

Table 2. Frequency of gluten exposure among adolescent participants of the Virtual Celiac Symptoms Study (2022–2023) during the 12-week study period stratified by level of adherence to a GFD at baseline.

	Overall	Self-reported level of adherence to a GFD at baseline: I eat gluten-containing foods regularly	I eat a GFD and rarely eat gluten on purpose	I eat a GFD and rarely eat gluten accidentally	I eat a GFD and never eat gluten accidentally or on purpose	*p* value
Number of participants, n (%)	142 (100)	3 (2.1)	9 (6.3)	111 (78.2)	19 (13.4)	–
Number of participants with known or suspected gluten exposure, n (%)	103 (72.5)	2 (66.7)	9 (100)	80 (72.1)	12 (63.2)	–
Frequency of gluten exposure (known or suspected), mean (SD) number of days during the 12-week study period	6.7 (8.7)	37.5 (38.9)	14.9 (7.0)	5.3 (6.1)	4.8 (4.5)	< 0.0001

GFD, gluten-free diet; SD, standard deviation.

Figure 1. Number of days with core CeD symptoms during the 12-week study period among adolescent participants (N = 142) of the Virtual Celiac Symptoms Study (2022–2023) who experienced each symptom.

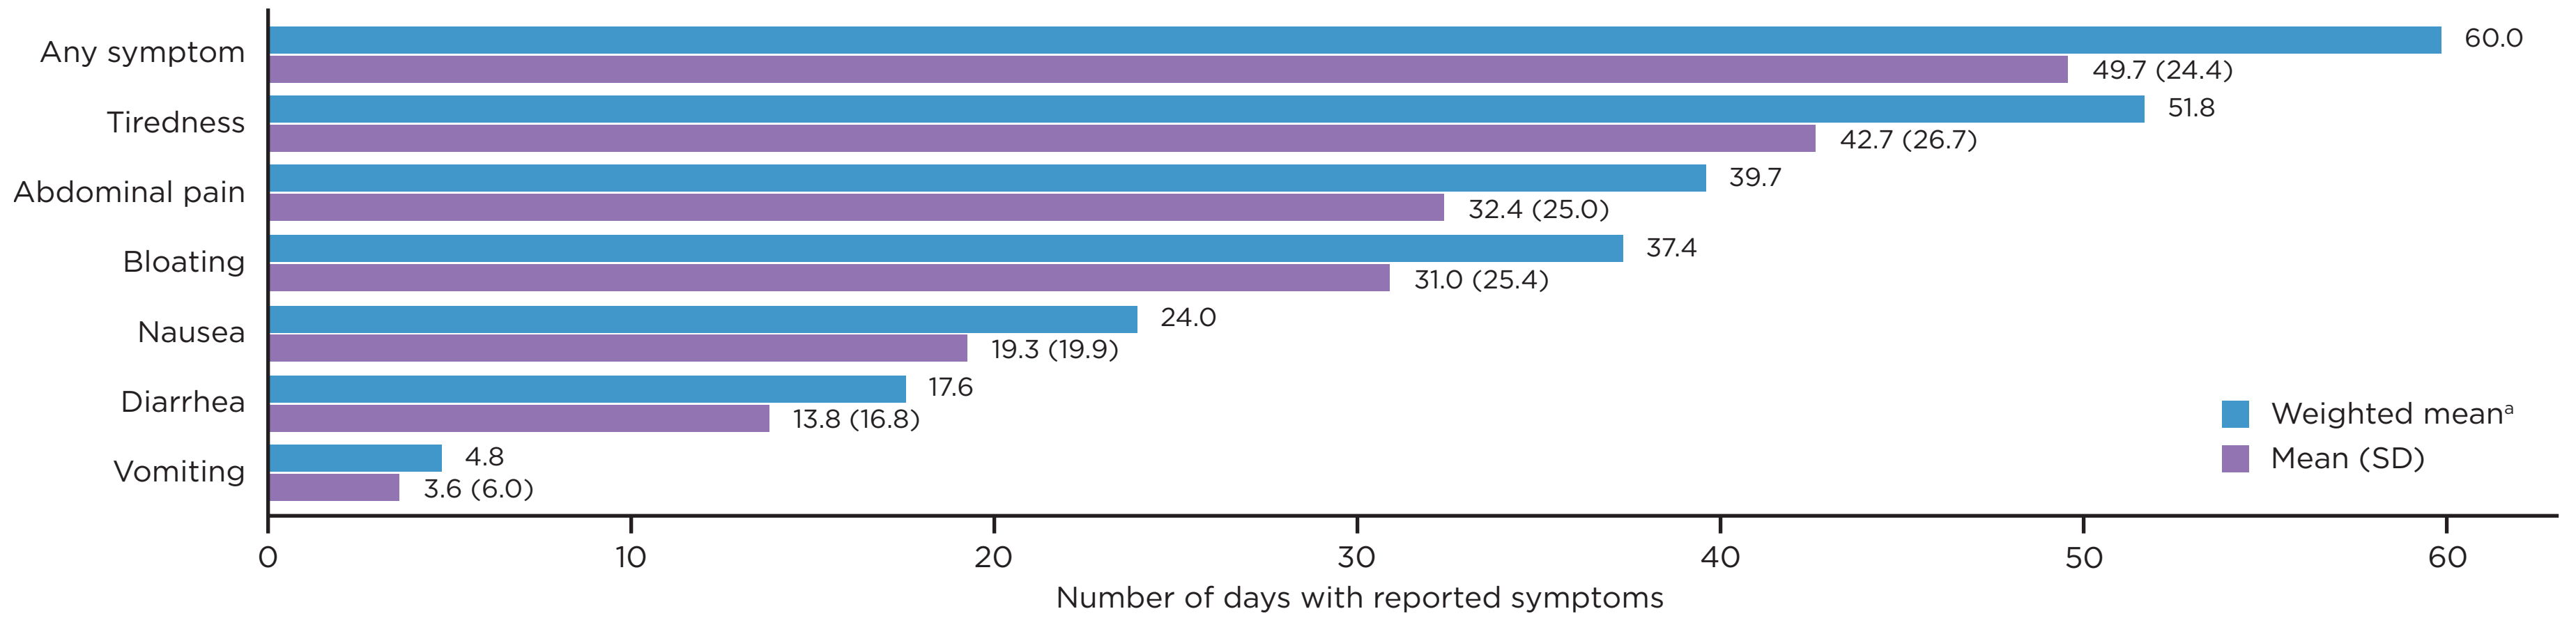
Any symptom
Tiredness
Abdominal pain
Bloating
Nausea
Diarrhea
Vomiting
60.0
49.7 (24.4)
51.8
42.7 (26.7)
39.7
32.4 (25.0)
37.4
31.0 (25.4)
24.0
19.3 (19.9)
17.6
13.8 (16.8)
4.8
3.6 (6.0)
Weighted mean[a]
Mean (SD)
0
10
20
30
40
50
60
Number of days with reported symptoms

[a]The weighted mean was calculated as the mean proportion of days with reported symptom(s) across participants weighted by the total study period days (84 days).
Symptoms were assessed using CDSD 2.1 and included diarrhea, abdominal pain, bloating, nausea, tiredness and vomiting. All participants (N = 142) with available data on ≥ 1 day were included in this analysis.
CDSD, Celiac Disease Symptom Diary; CeD, celiac disease; SD, standard deviation.

Figure 2. CeD core symptoms experienced by adolescent participants during the 12-week study period stratified by level of adherence to a GFD self-reported at baseline.

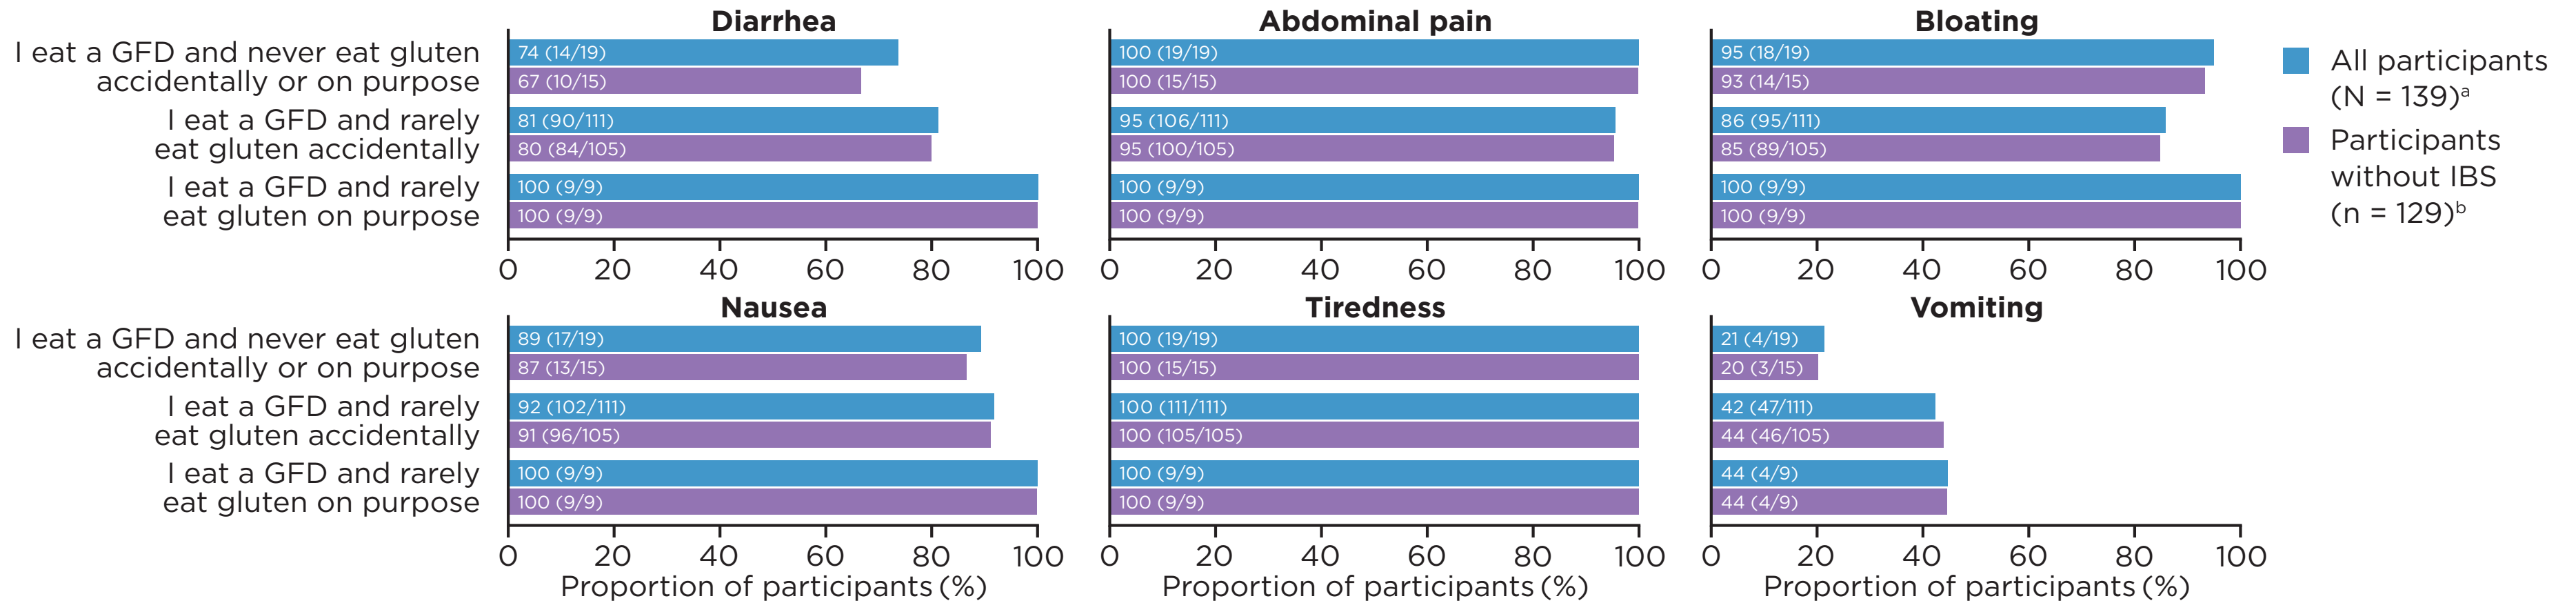
I eat a GFD and never eat gluten accidentally or on purpose
I eat a GFD and rarely eat gluten accidentally
I eat a GFD and rarely eat gluten on purpose
Diarrhea
74 (14/19)
67 (10/15)
81 (90/111)
80 (84/105)
100 (9/9)
100 (9/9)
Abdominal pain
100 (19/19)
100 (15/15)
95 (106/111)
95 (100/105)
100 (9/9)
100 (9/9)
Bloating
95 (18/19)
93 (14/15)
86 (95/111)
85 (89/105)
100 (9/9)
100 (9/9)
Nausea
89 (17/19)
87 (13/15)
92 (102/111)
91 (96/105)
100 (9/9)
100 (9/9)
Tiredness
100 (19/19)
100 (15/15)
100 (111/111)
100 (105/105)
100 (9/9)
100 (9/9)
Vomiting
21 (4/19)
20 (3/15)
42 (47/111)
44 (46/105)
44 (4/9)
44 (4/9)
0
20
40
60
80
100
Proportion of participants (%)
All participants (N = 139)[a]
Participants without IBS (n = 129)[b]

[a]Total number of adolescent participants was 139 because none of the participants reported "*eating gluten-free diet sometimes*" and participants who reported "*eating gluten-containing foods regularly*" (n = 3) were not included in this analysis.
[b]Total number of adolescent participants was 129 because participants who reported "*eating gluten-containing foods regularly*" (n = 2) were not included in this analysis.
CeD, celiac disease; GFD, gluten-free diet; IBS, irritable bowel syndrome. Data inside bars represent % (n/N).

- Among the core CeD symptoms experienced by adolescent participants during the 12 weeks of the study, tiredness, abdominal pain and bloating had the highest number of days with symptom occurrence (**Figure 1**).
- All adolescent participants reported at least one core CeD symptom.
 - Each GI symptom occurred at least once in > 80% of adolescent participants (with the exception of diarrhea in participants who reported adhering to a GFD and "*never eating gluten accidentally or on purpose*" and vomiting; **Figure 2**).
 - Adolescent participants experienced symptoms regardless of level of adherence to a GFD (self-reported at baseline) or IBS diagnosis (**Figure 2**).

Limitations

- There is currently no classification of symptom severity for CeD, thus participant self-assessment was used for this study.
- The self-reporting of daily symptoms and dietary intake are subject to inherent biases. Furthermore, daily reporting could increase awareness of GFD adherence, which may lead to changes in participants' dietary choices during the course of the study.
- There may be misclassification of celiac diagnosis status by participants.
- The experience among adolescent patients with CeD in countries other than the USA may differ.

Acknowledgments

The authors would like to acknowledge Lin Zou, Allison Quintana and Mariel Arvizu for analytical support, Sanjana Sundaresan and James Signorovitch for consulting services and Julia McBeth and Marissa Mahoney for study logistics support.

Conclusions

- **Despite self-reported adherence to a GFD, all adolescent participants reported at least one GI symptom.**
- **The findings from this study emphasize the need to better delineate between symptoms perceived to be related to gluten exposure and those that are not, and to develop therapies for the treatment and management of CeD among adolescent patients on a GFD.**
- **Symptom patterns identified in this study indicate that specific GI symptoms could be explored as relevant endpoints in future research of potential CeD medications.**

References

1. Caio G *et al. BMC Med* 2019;17:142.
2. Leffler DA *et al. Value Health* 2017;20:637–43.
3. Rubio-Tapia A *et al. Am J Gastroenterol* 2023;118:59–76.
4. Hall NJ *et al. Appetite* 2013;68:56–62.
5. Leffler DA *et al. Dig Dis Sci* 2008;53:1573–81.
6. Singh J *et al. J Hum Nutr Diet* 2011;24:479–86.
7. Syage JA *et al. Am J Clin Nutr* 2018;107:201–7.
8. Chang D *et al. Aliment Pharmacol Ther* 2022; 56 Suppl 1:s73–85.
9. Mapi Research Trust. Available from: https://eprovide.mapi-trust.org. (Accessed September 2023).

Disclosures

DA and **EL** serve as consultants for Takeda Pharmaceuticals. **LMM** was an employee of Takeda Development Center Americas, Inc. at the time of this study and holds Takeda stock. **DAL** is an employee of Takeda Development Center Americas, Inc. and receives stock or stock options. **JRM**, **SS** and **ES** are employees of Analysis Group, Inc., which received research support from Takeda Development Center Americas, Inc. **MG** is an employee of the Celiac Disease Foundation, which received financial support from Takeda Development Center Americas, Inc.

Funding

This study was sponsored by Takeda Development Center Americas, Inc. Medical writing support was provided by Christina Nikolakopoulou, PhD, of Oxford PharmaGenesis, Oxford, UK and was funded by Takeda Pharmaceuticals.

Presented at the North American Society for Pediatric Gastroenterology, Hepatology and Nutrition Annual Meeting, October 4–7, 2023, San Diego, CA

Corresponding author: Edwin.Liu@childrenscolorado.org